AASLD The Liver Meeting®

DIGITAL IMAGE QUANTIFICATION OF COLLAGEN PROPORTIONATE AREA CORRELATES WITH METAVIR FIBROSIS STAGE AND LIVER STIFFNESS MEASUREMENT VIA FIBROSCAN® IN ALPHA-1 ANTITRYPSIN DEFICIENCY-ASSOCIATED LIVER DISEASE

V. GUPTA[1], F. HONG[1], J.-C. CHUANG[1], D. CORDOVER[1], J. CHENG[1], N.K. DESAI[1], S. GONZALEZ[1], P. THAKKER[1], T. SCHLUEP[2], P. STRNAD[3], V.C. CLARK[4], R. LOOMBA[5], R. SAXENA[6], C. BEHLING[5] and K. WASHINGTON[7]

[1]Takeda Development Center Americas, Inc., Cambridge, MA, USA; [2]Arrowhead Pharmaceuticals, Inc., Pasadena, CA, USA; [3]University Hospital RWTH Aachen, Aachen, Germany; [4]University of Florida, Gainesville, FL, USA; [5]University of California San Diego, San Diego, CA, USA; [6]Emory University School of Medicine, Atlanta, GA, USA; [7]Vanderbilt University Medical Center, Nashville, TN, USA

Poster 4454

Please scan the QR code to download a copy of the poster.
Alternatively, please use the following link: https://tiny.one/5o116707w9
Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from AASLD and the authors

INTRODUCTION

- Alpha-1 antitrypsin deficiency (AATD) is a rare genetic disease that may lead to the development of liver and/or lung disease.[1]
- The most severe AATD phenotypes are associated with the protease inhibitor (Pi)*ZZ genotype, which causes the accumulation of misfolded alpha-1 antitrypsin (Z-AAT) in hepatocytes and increased risk of developing AATD-associated liver disease (AATD-LD).[1]
- Currently, there are no approved pharmacological therapies available for AATD-LD; consequently, liver transplantation is recommended in patients with AATD and advanced liver cirrhosis or failure.[2]
- Fazirsiran is an investigational small interfering RNA therapy undergoing phase 3 development in patients with AATD-LD.[3,4]
- Liver biopsy is the reference standard for the assessment of fibrosis in liver diseases.[5]
- Severity of liver fibrosis is determined by pathologist-led Meta-analysis of Histological Data in Viral Hepatitis (METAVIR) fibrosis stage.[6]
- Change in liver fibrosis is an important endpoint for the evaluation of efficacy of investigational therapies.
 - However, pathologist-reported histology evaluation provides categorical scores which may be influenced by inter- and intra-reader variability.[7]
 - Therefore, improved methods to objectively evaluate histological changes are needed.
- Digital quantification by collagen proportionate area (CPA) offers a reproducible and continuous score, capable of capturing dynamic changes in fibrosis.[8]

AIM

- To establish a digital quantification method for CPA in AATD-LD and compare it with METAVIR fibrosis stage and FibroScan® scores using data from phase 2 clinical trials of fazirsiran.

METHOD

Figure 1. CPA method

CPA method was established using HALO™ and HALO AI (Indica Labs, Albuquerque, NM, USA)

PSR staining → AI-based tissue segmentation delineates collagen, hepatocytes, nuclei, vessels, glands and artifacts (HALO AI DenseNet2 Classifier) → Area quantification algorithm within collagen class (HALO Area Quantification v2.4.3) → Data output

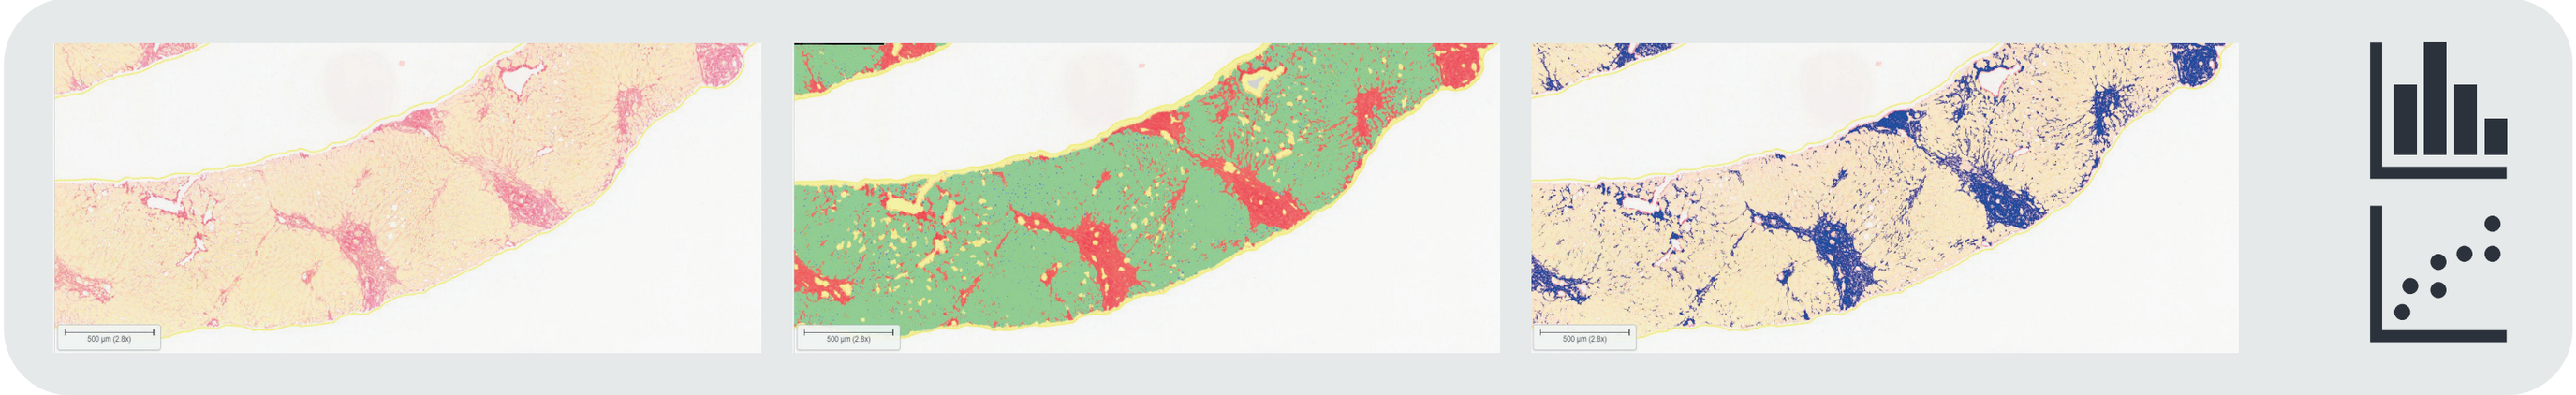

- Samples from patients with AATD-LD who received fazirsiran/placebo and were enrolled in the AROAAT-2001 (NCT03945292) and AROAAT-2002 (NCT03946449) phase 2 trials were included in the analysis.[3,4]
- Formalin-fixed, paraffin-embedded, archived liver biopsy samples were utilized to prepare 4 μM-thick sections for picrosirius red (PSR) staining using a commercially available Abcam kit (ref: ab150681, Abcam, USA) by a central laboratory; slides were scanned using a Leica AT2 scanner (Leica Biosystems, UK) at 40× magnification.
- For CPA analysis, a DenseNet machine-learning algorithm was trained to identify different tissue morphologies for detection of fibrotic collagen, hepatocytes, nuclei, vessels and glands; collagen was quantified within fibrotic collagen using the area quantification method in whole-slide images of PSR-stained liver biopsy sections.
- METAVIR fibrosis stage was centrally read using semi-quantitative scales and adjudicated by three hepatopathologists blinded to treatment, patient identification and time point, and published previously.[3,4]
- Correlations of digitally quantified CPA with pathologist-reported METAVIR fibrosis stage and liver stiffness measurement via FibroScan were evaluated by Spearman rank correlation analysis.
- METAVIR fibrosis staging, FibroScan and additional methodological details of the previous phase 2 trials were described by Clark *et al.* in 2024 and Strnad *et al.* in 2022.[3,4]

RESULTS

Table 1. Baseline patient characteristics

Overall, 41 patients from the AROAAT-2001 (*n* = 25) and AROAAT-2002 (*n* = 16) trials had evaluable paired baseline and post-treatment liver histological data

	AROAAT-2001 (*n* = 25)	AROAAT-2002 (*n* = 16)	Overall (*N* = 41)
Age, years, median (IQR)	57.0 (47.0, 64.0)	56.0 (49.0, 62.2)	56.0 (47.0, 63.0)
BMI, kg/m^2, median (IQR)	28.9 (24.8, 36.1)	25.1 (21.8, 27.9)	26.6 (24.4, 31.2)
Male, *n* (%)	14 (56.0)	14 (87.5)	28 (68.3)
METAVIR fibrosis stage, *n* (%)			
0	3 (12.0)	1 (6.3)	4 (9.8)
1	9 (36.0)	2 (12.5)	11 (26.8)
2	11 (44.0)	6 (37.5)	17 (41.5)
3	2 (8.0)	4 (25.0)	6 (14.6)
4	0	2 (12.5)	2 (4.9)
Missing	0	1 (6.3)	1 (2.4)
CPA, %, median (IQR)	2.9 (1.7, 5.1)	2.3 (1.6, 4.2)	2.6 (1.7, 4.6)
Liver stiffness measurement via FibroScan, kPA, median (IQR)	7.6 (4.4, 11.0)	9.2 (7.1, 11.8)	7.9 (5.6, 11.4)

Figure 3. Cross-sectional analysis (baseline and post-treatment) of the correlation between CPA and METAVIR fibrosis stage and between CPA and liver stiffness measurement via FibroScan

CPA correlated significantly with METAVIR fibrosis stage (ρ = 0.41, *p* = 0.0002) and liver stiffness measurement via FibroScan (ρ = 0.28, *p* = 0.02)

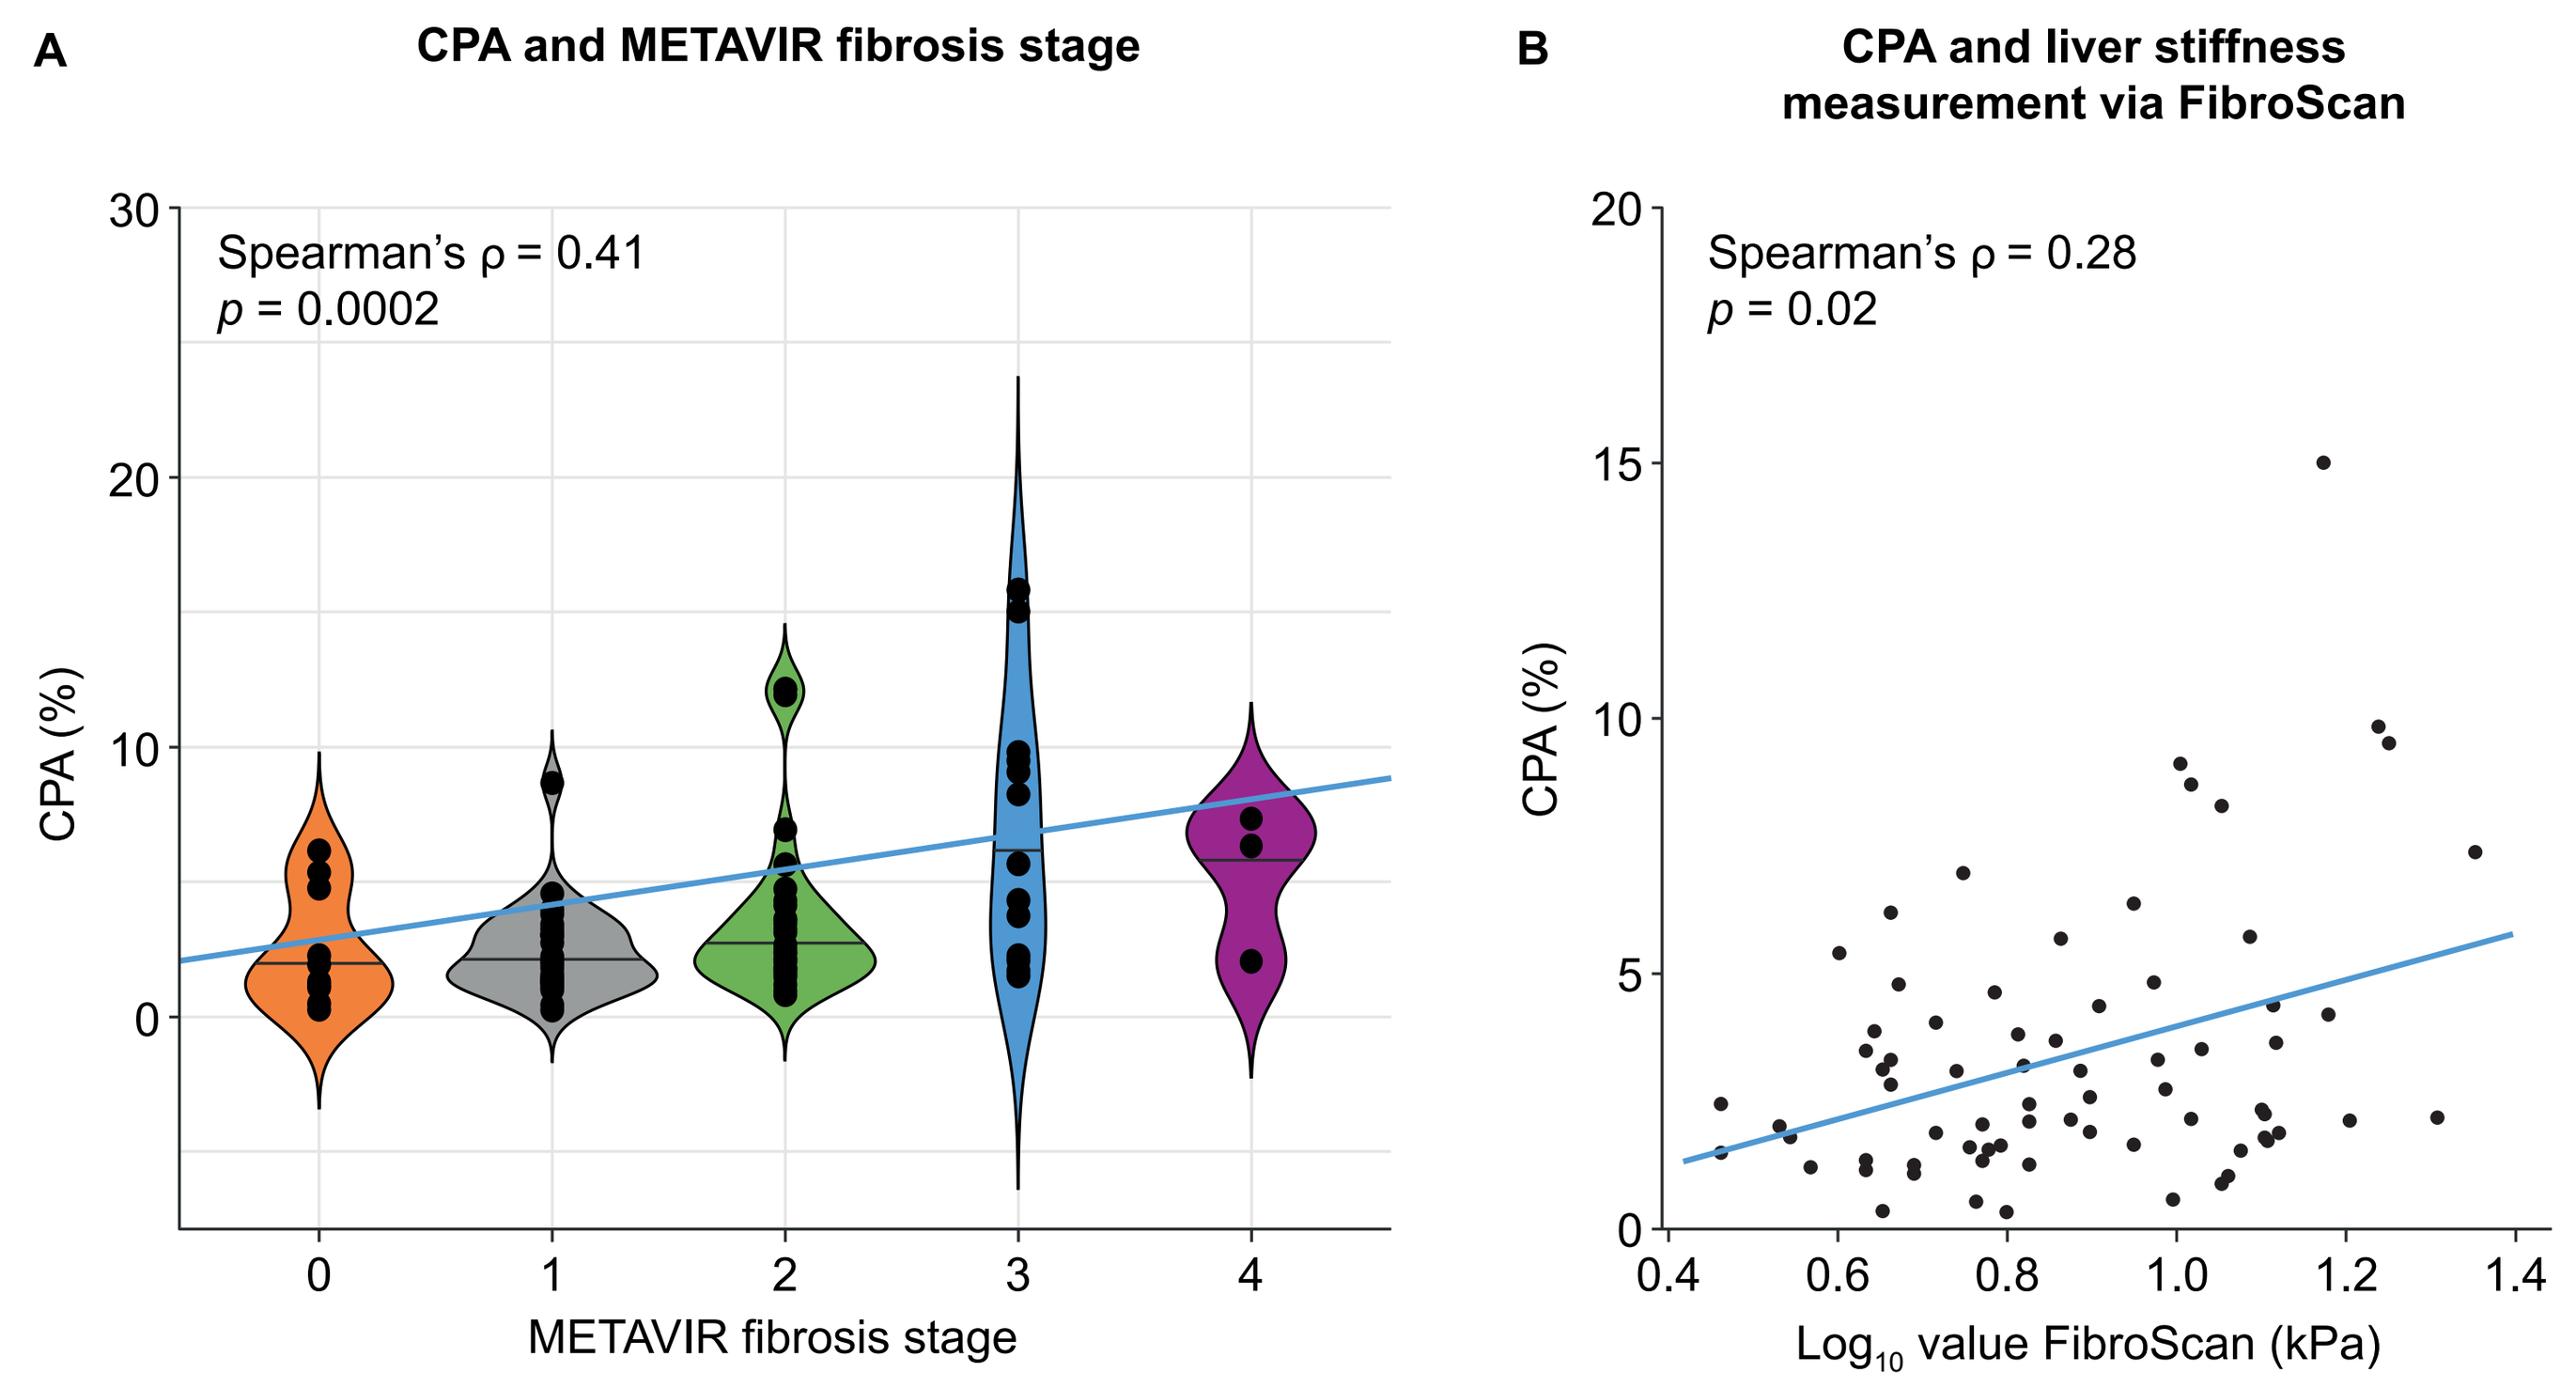

A
CPA and METAVIR fibrosis stage
Spearman's ρ = 0.41
p = 0.0002
CPA (%)
METAVIR fibrosis stage
B
CPA and liver stiffness measurement via FibroScan
Spearman's ρ = 0.28
p = 0.02
CPA (%)
Log10 value FibroScan (kPa)

Figure 4. Correlation of the change in CPA, liver stiffness measurement via FibroScan and METAVIR fibrosis stage

Change in CPA was significantly correlated with change in liver stiffness measurement via FibroScan post-treatment (Figure 4A)

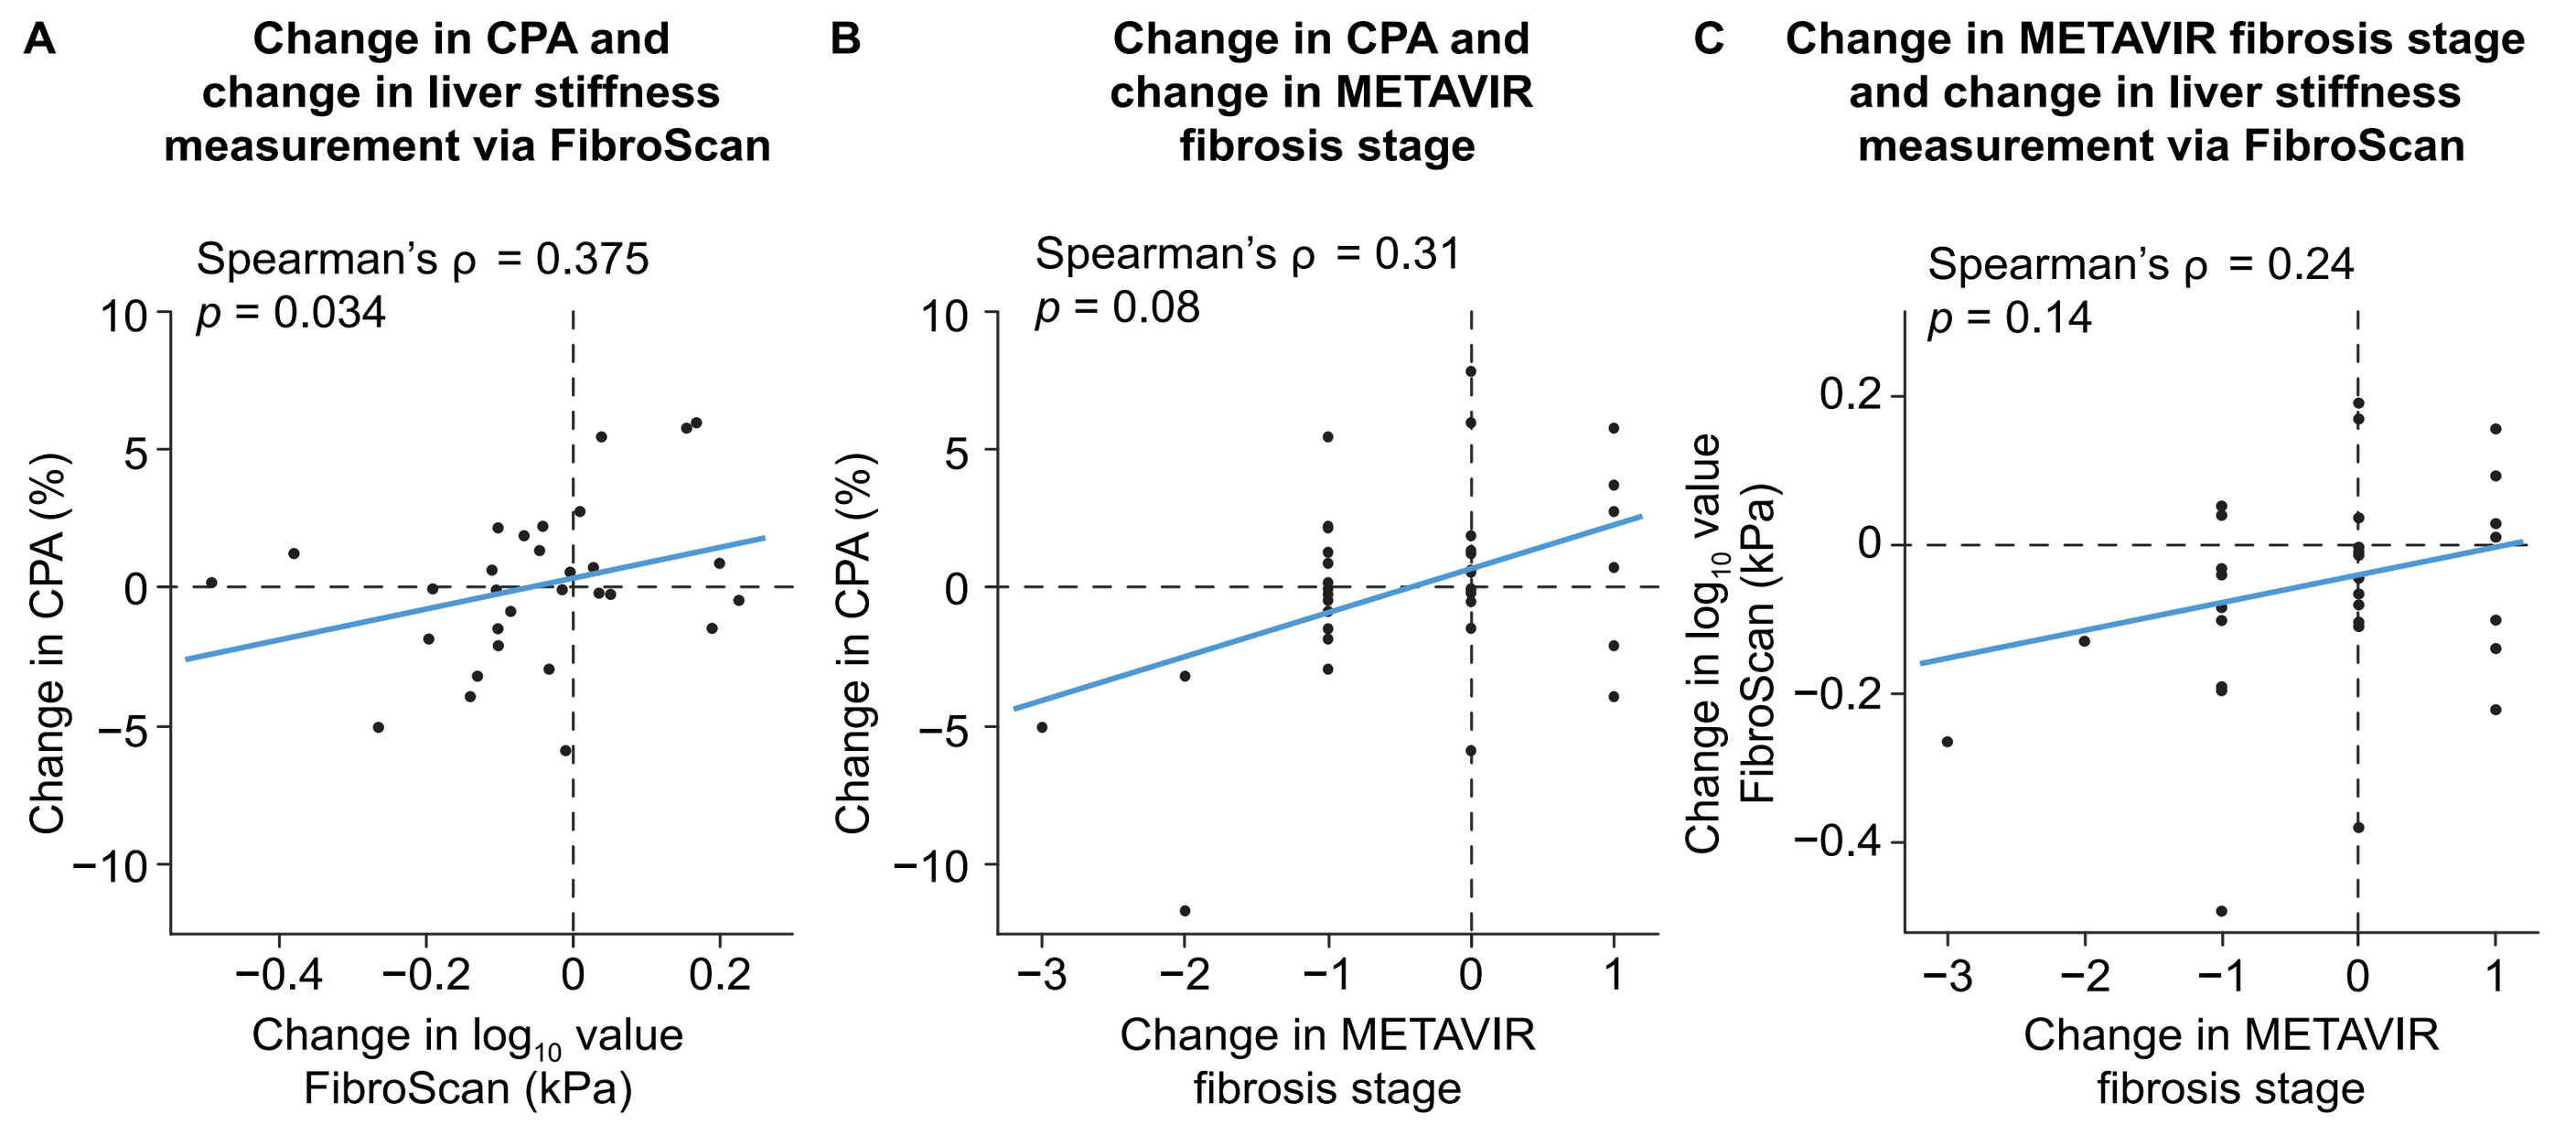

A
Change in CPA and change in liver stiffness measurement via FibroScan
Spearman's ρ = 0.375
p = 0.034
Change in CPA (%)
Change in log10 value FibroScan (kPa)
B
Change in CPA and change in METAVIR fibrosis stage
Spearman's ρ = 0.31
p = 0.08
Change in CPA (%)
Change in METAVIR fibrosis stage
C
Change in METAVIR fibrosis stage and change in liver stiffness measurement via FibroScan
Spearman's ρ = 0.24
p = 0.14
Change in log10 value FibroScan (kPa)
Change in METAVIR fibrosis stage

- Change in CPA showed a positive but not statistically significant correlation with change in METAVIR fibrosis stage post-treatment (**Figure 4B**).
- Change in METAVIR fibrosis stage showed a positive but not statistically significant correlation with change in liver stiffness measurement via FibroScan post-treatment (**Figure 4C**).

Figure 2. Pre- and post-treatment PSR-stained biopsies and markup for CPA digital quantification

Representative images were taken from two patients with progression or regression of liver fibrosis

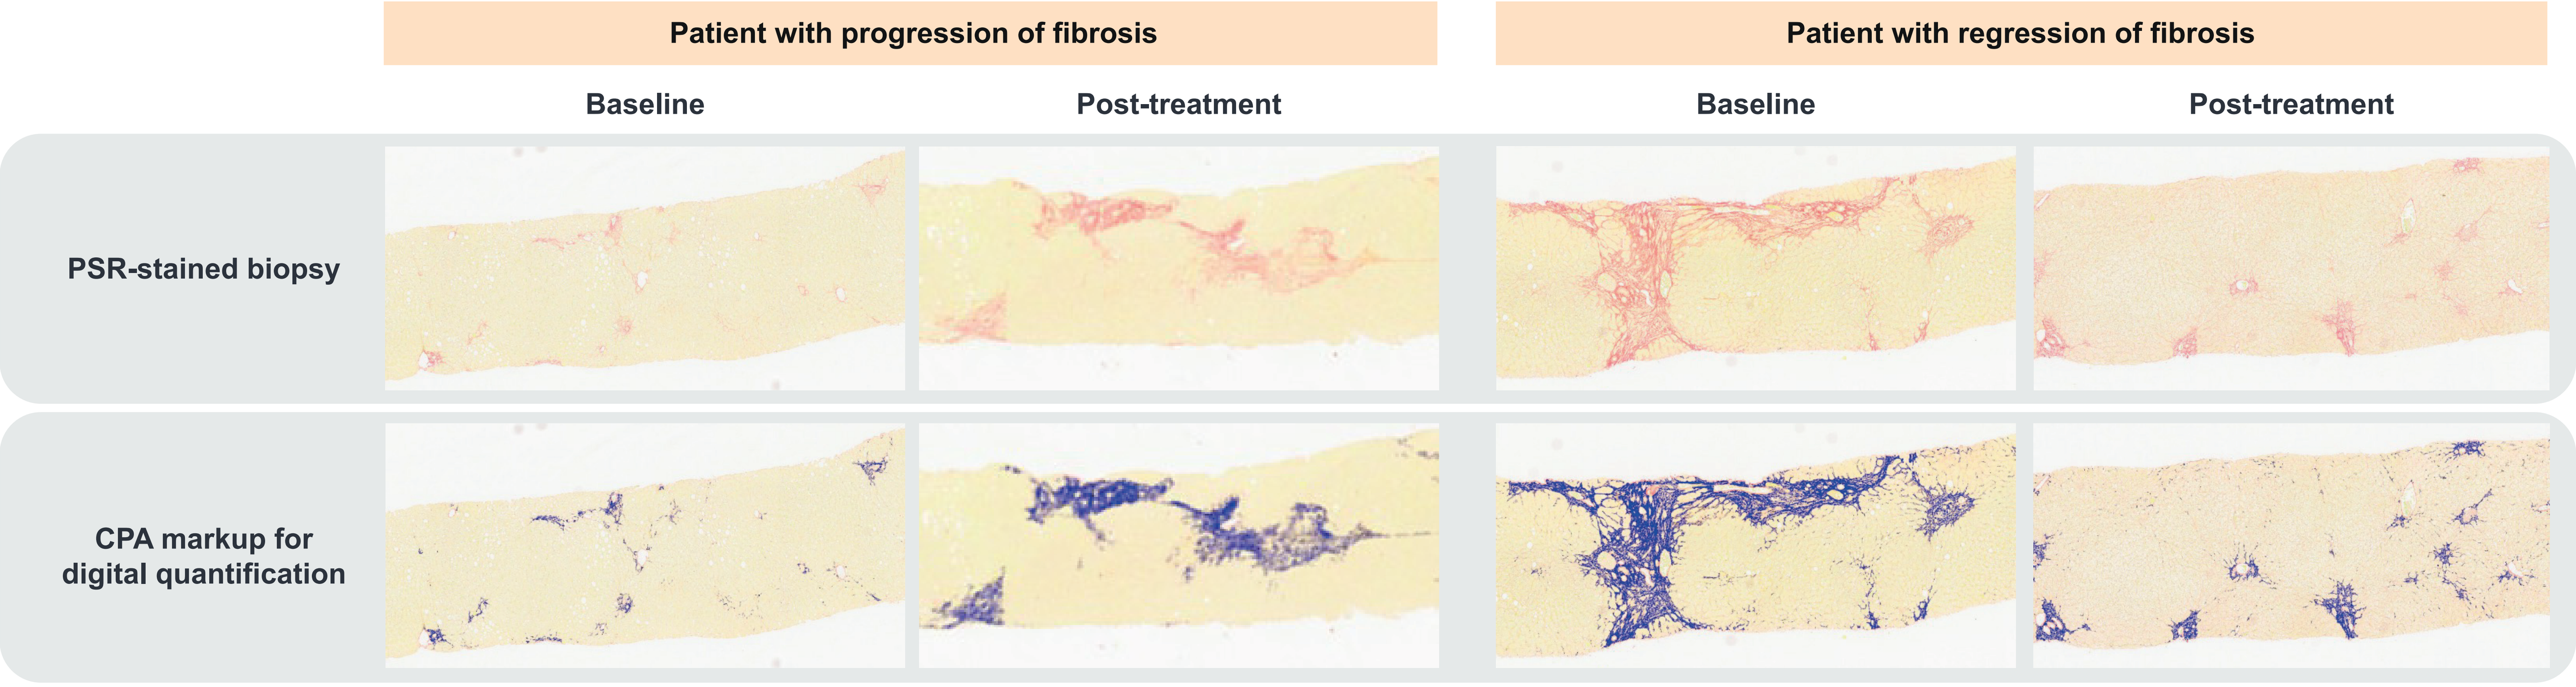

Patient with progression of fibrosis
Patient with regression of fibrosis
Baseline
Post-treatment
Baseline
Post-treatment
PSR-stained biopsy
CPA markup for digital quantification

CONCLUSIONS AND FUTURE DIRECTIONS

- CPA correlated significantly with METAVIR fibrosis stage and liver stiffness measurement via FibroScan.
 - This suggests value in continuous scores for the quantification of liver fibrosis and potential for the utility of CPA as a measure of fibrosis in AATD-LD biopsy to complement pathologist-led histological evaluation.
- Comparison with other noninvasive tests, including magnetic resonance elastography and Fibrosis-4, should be considered where data are available.
- More advanced AI/machine-learning approaches may further substantiate analyses and support therapeutic development in AATD-LD.

LIMITATIONS

- The sample size from the combined AROAAT-2001 and AROAAT-2002 trials was small (*n* = 41) and most patients had a low METAVIR fibrosis stage (≤ 2).
- Both CPA and METAVIR fibrosis staging are biopsy-based assessments and may be affected by sample size, location, quality and staining; METAVIR fibrosis stage may also be influenced by inter- and intra-reader variability.
- CPA is a continuous score derived by automated methods but does not capture architectural changes.

ABBREVIATIONS
AATD, alpha-1 antitrypsin deficiency; AATD-LD, alpha-1 antitrypsin deficiency-associated liver disease; AI, artificial intelligence; BMI, body mass index; CPA, collagen proportionate area; IQR, interquartile range; METAVIR, Meta-analysis of Histological Data in Viral Hepatitis; PSR, picrosirius red; Z-AAT, misfolded alpha-1 antitrypsin.

ACKNOWLEDGMENTS
John Dahlquist, William Webber and Abishek Chandrashekar from Takeda made significant contributions by facilitating samples and image management. Support from Indica Labs in digital pathology and Geneuity for PSR staining is appreciated. At the direction of the authors, medical writing assistance was provided by Rebecca Tooze, PhD, of Oxford PharmaGenesis, Oxford, UK.

FUNDING
This study and medical writing support was funded by Takeda Development Center Americas, Inc.

REFERENCES

1. Strnad P *et al. N Engl J Med* 2020;382:1443–55.
2. Patel D, Teckman J. *Ther Adv Chronic Dis* 2021;12 (Suppl): 2040622321995684.
3. Clark VC *et al. Gastroenterology* 2024. doi: 10.1053/j.gastro.2024.06.028 (online ahead of print).
4. Strnad P *et al. N Engl J Med* 2022;387:514–24.
5. Fromme M *et al. J Hepatol* 2022;76:946–58.
6. Chowdhury AB, Mehta KJ. *Clin Exp Med* 2023;23:273–85.
7. Sebastiani G. *World J Gastroenterol* 2009;15:2190–203.
8. Gawrieh S *et al. Ann Diagn Pathol* 2020;47:151518.

DISCLOSURES
VG, **FH**, **J-CC**, **DC**, **JC**, **NKD**, **SG** and **PT** are employees and stockholders of Takeda Development Center Americas, Inc. **TS** is an employee of Arrowhead Pharmaceuticals. **PS** has received grant support and lecture fees from CSL Behring; grant support from Arrowhead Pharmaceuticals, Dicerna Pharmaceuticals and Vertex Pharmaceuticals; advisory board/consulting fees from BioMarin Pharmaceuticals, BridgeBio, GlaxoSmithKline, Intellia Pharmaceuticals, Ipsen Pharmaceuticals, Novo Nordisk, Swedish Orphan Biovitrum AB and Takeda Pharmaceuticals. **VCC** has received research support from Novo Nordisk, Takeda and Vertex; and consulting fees from Takeda and Vertex. **RL** serves as a consultant to 89bio, Aardvark Therapeutics, Alnylam/Regeneron, Altimmune, Amgen, Arrowhead Pharmaceuticals, AstraZeneca, Bristol Myers Squibb, CohBar, Eli Lilly, Galmed Pharmaceuticals, Gilead, Glympse Bio, HighTide, Inipharma, Intercept, Inventiva, Ionis, Janssen, Inc., Madrigal Pharmaceuticals, Merck, Metacrine, Inc., NGM Biopharmaceuticals, Novartis, Novo Nordisk, Pfizer, Sagimet Biosciences, Terns Pharmaceuticals, Theratechnologies and Viking Therapeutics. He has stock options in 89bio and Sagimet Biosciences. In addition, his institutions have received research grants from Arrowhead Pharmaceuticals, AstraZeneca, Boehringer Ingelheim, Bristol Myers Squibb, Eli Lilly, Galectin Therapeutics, Galmed Pharmaceuticals, Gilead, Hanmi, Intercept, Inventiva, Ionis, Janssen, Inc., Madrigal Pharmaceuticals, Merck, NGM Biopharmaceuticals, Novo Nordisk, Pfizer, Sonic Incytes and Terns Pharmaceuticals. He is also the co-founder of LipoNexus, Inc. **RS** serves as a consultant to Takeda. **CB** serves as a consultant with Biomarin Pharmaceuticals and Pathology Institute; and provides service-related contract work for Akero, Hanmi (through Labcorp) and Novo Nordisk. **KW** serves as a consultant to Takeda.

Presented at the American Association for the Study of Liver Diseases (AASLD) Liver Meeting 2024; November 15–19, 2024, San Diego, CA, USA