Proteomic analysis identified fazirsiran treatment-responsive protein biomarkers in patients with alpha-1 antitrypsin deficiency-associated liver disease

Jen-Chieh Chuang,[1] Ruixue Hou,[1] Feng Hong,[1] Jie Cheng,[1] Maria D Paraskevopoulou,[1] Nirav K Desai,[1] Paresh Thakker,[1] Thomas Schluep,[2] Pavel Strnad[3]

[1]Takeda Development Center Americas, Inc., Cambridge, MA, USA; [2]Arrowhead Pharmaceuticals, Inc., Pasadena, CA, USA; [3]University Hospital RWTH Aachen, Aachen, Germany

Poster FRI-164

Please scan the QR code to download a copy of the poster and supplementary material. Alternatively, please use the following link: https://tiny.one/8dD1454MSH. Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from EASL and the authors.

Introduction

- Alpha-1 antitrypsin deficiency (AATD) is a rare genetic disease characterized by low levels of serum alpha-1 antitrypsin (AAT), which primarily affects the lungs and/or liver.[1]
- Patients with the protease inhibitor (Pi)*ZZ genotype express misfolded AAT (Z-AAT), resulting in hepatic Z-AAT aggregates and reduced antiprotease activity in the lungs.[1,2]
- Accumulation of proteotoxic Z-AAT polymers leads to hepatic cell stress, inflammation and cell damage, and liver fibrosis with increased extracellular matrix turnover.[3]
- Currently no pharmacological treatments for AATD-associated liver disease (AATD-LD) exist.[4]
 - Fazirsiran is an investigational small interfering RNA undergoing phase 3 development in patients with AATD-LD (NCT05677971).
- Proteomics data to identify biomarkers relevant for therapeutic targeting and to elucidate disease pathophysiology are limited.

Objective

- To detect treatment-responsive protein biomarkers and cellular pathways by leveraging a novel proteomic platform for biomarker discovery using serum samples from patients with AATD-LD treated with fazirsiran.

Methods

- Olink Explore 3072, a high-throughput dual-antibody-based proteomics platform, was utilized for protein biomarker discovery.
- Serum samples were assessed at baseline and at 4, 16, 24, 28 and 48 weeks post-treatment initiation from 16 adults with AATD-LD, a Pi*ZZ genotype, and biopsy-proven liver fibrosis who participated in AROAAT-2002 (NCT03946449), a phase 2, open-label trial of fazirsiran (100 or 200 mg); treatment with fazirsiran resulted in > 80% reduction in serum and liver Z-AAT.[5]
 - Additional study design information has been previously described.[5]
- Olink data were integrated with single nucleus RNA-sequencing (Snucseq) data to map expressed proteins to potential source cells in the liver and improve data interpretability.
 - The Snucseq data consisted of samples from four patients with non-alcoholic steatohepatitis (NASH)/metabolic dysfunction-associated steatohepatitis (MASH) and two healthy controls.
- A mixed effects model was applied to measure biomarker change from baseline over time and a false discovery rate (FDR)-adjusted p-value was used to select the top biomarkers (**Figure 1**).
- Differentially expressed protein (DEP) and pathway analyses were also conducted.

Figure 1. Statistical and bioinformatic approaches identified fazirsiran-responsive serum protein markers

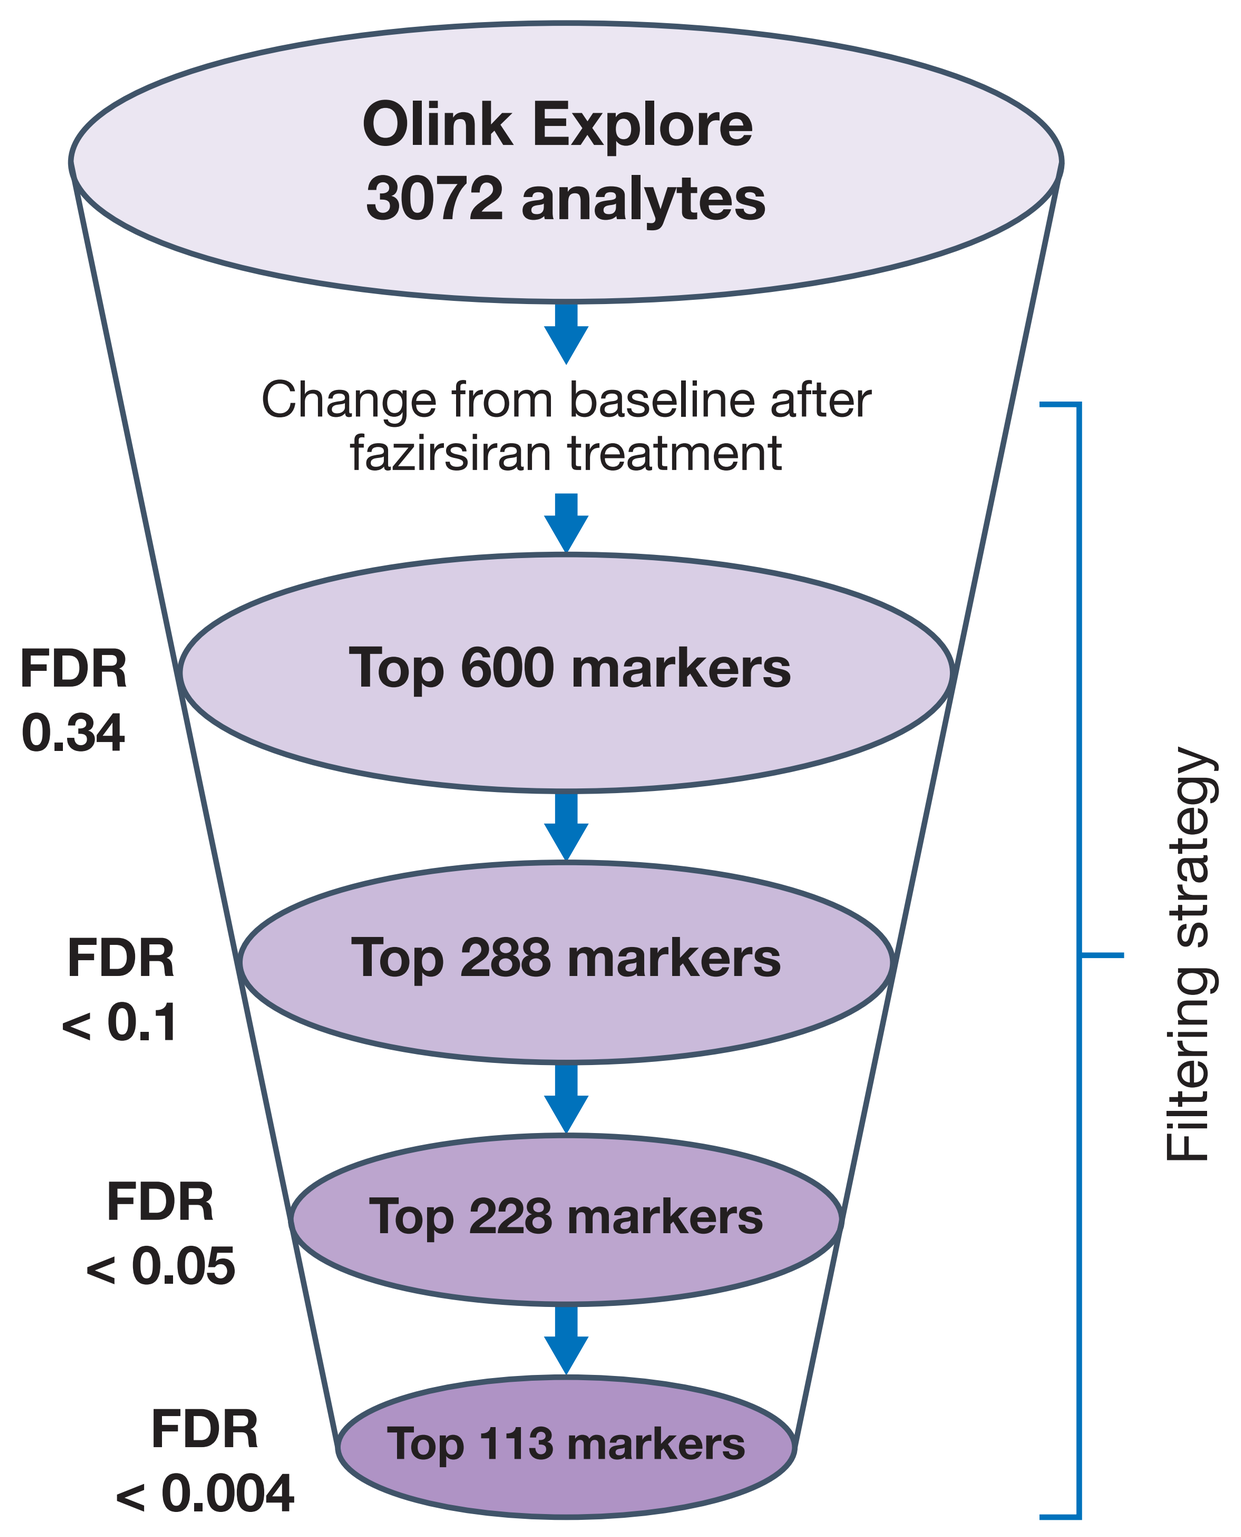
Olink Explore
3072 analytes
Change from baseline after fazirsiran treatment
FDR 0.34
Top 600 markers
FDR < 0.1
Top 288 markers
FDR < 0.05
Top 228 markers
FDR < 0.004
Top 113 markers
Filtering strategy

The Olink panel was integrated with Snucseq data to identify potential cell sources in the liver

A mixed effects model was used to assess biomarker change from baseline at all time points

FDR-adjusted p-values were used to filter the top biomarkers

Targeted filtering approach:
- Identify cell-type selective markers
- Identify markers based on existing knowledge/data in the literature

Systemic filtering approach:
- A pathway analysis was performed with an over-representation analysis to identify the top 113, 228, 288 and 600 markers

Results

- Integration of the Olink panel with the Snucseq data is shown in **Figure 2**.
- Fazirsiran treatment resulted in continuous and sustained reductions in DEPs in serum through 48 weeks (**Figure 3A**).
- DEPs were primarily mapped to hepatocytes, mesenchymal cells, immune cells (T cells and macrophages), cholangiocytes and endothelial cells (**Figure 3B**).
 - DEPs revealed potential common mechanisms associated with liver fibrosis between AATD-LD and other liver diseases, including NASH/MASH and non-alcoholic fatty liver disease/metabolic dysfunction-associated fatty liver disease.
 - Of the liver mesenchymal cell-enriched proteins, many were components of the extracellular matrix associated with hepatic stellate cell activation and liver fibrosis (**Figure 4**).
 - Downregulated proteins that mapped to hepatocytes were associated with cell stress and apoptosis.

Figure 2. Integration of the Olink panel with Snucseq data suggested liver cell sources for the circulating biomarkers

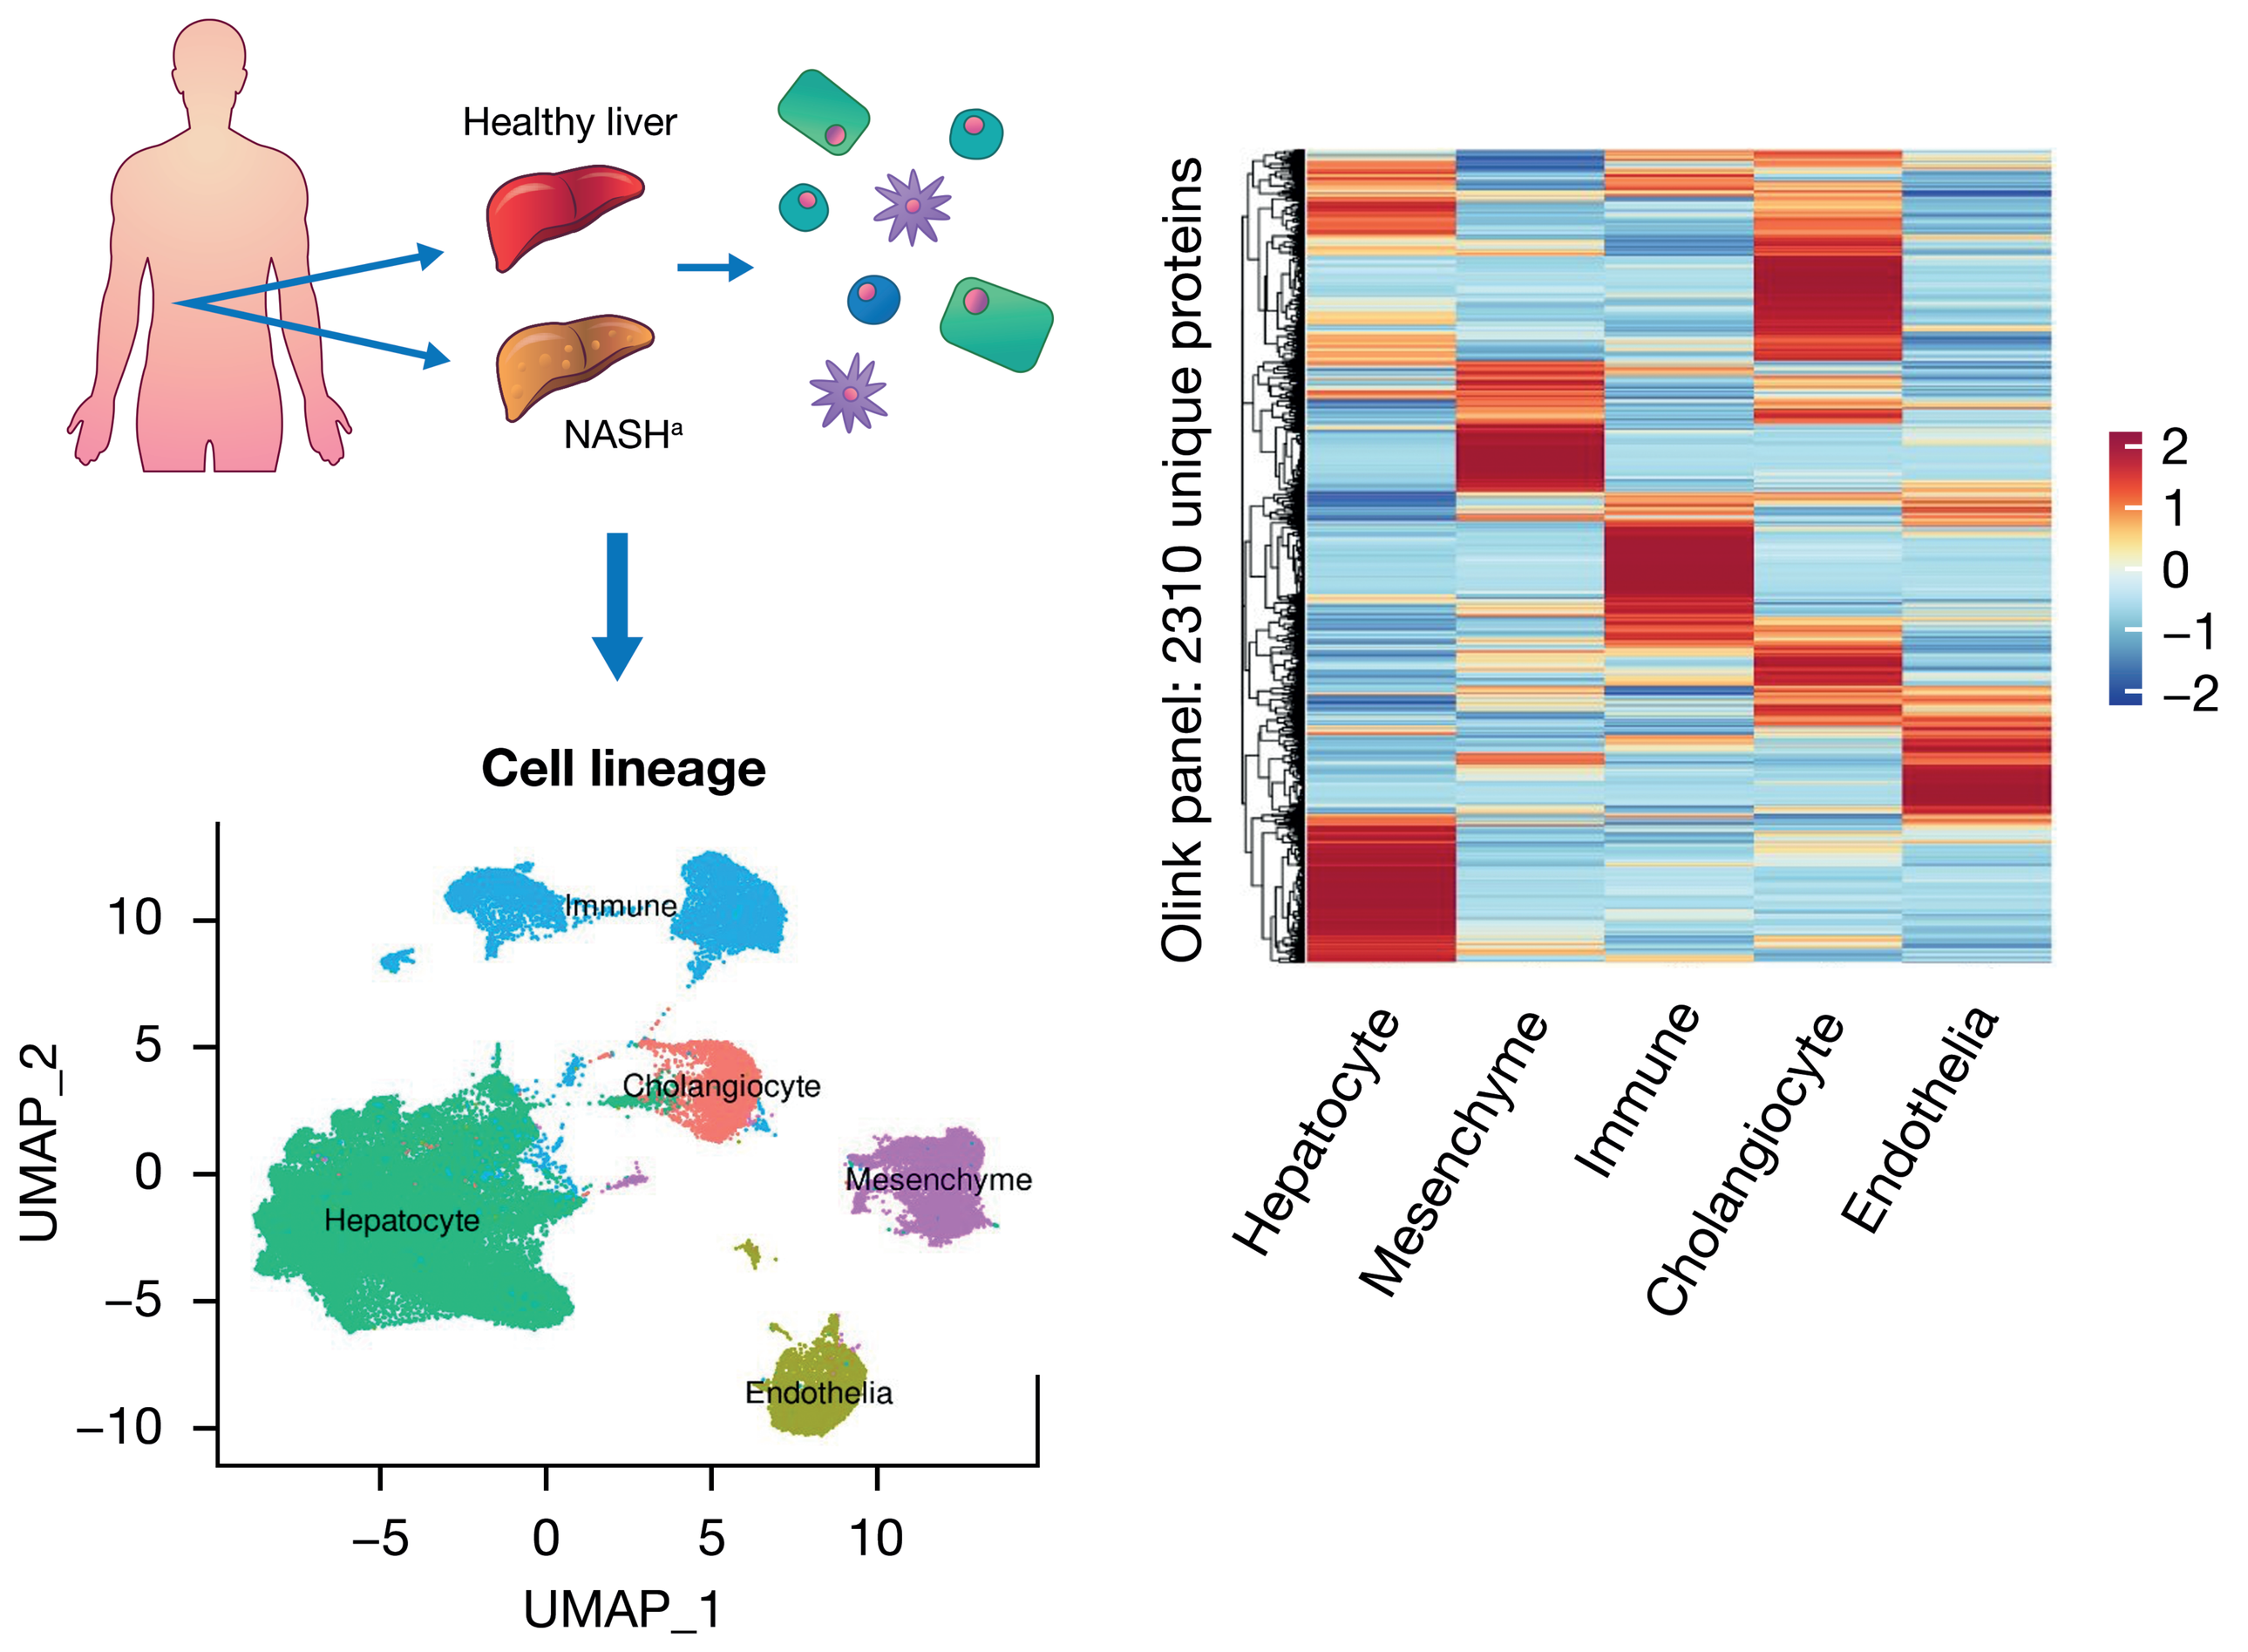
Healthy liver
NASH[a]
Cell lineage
Immune
Cholangiocyte
Hepatocyte
Mesenchyme
Endothelia
UMAP_2
UMAP_1
10
5
0
−5
−10
Olink panel: 2310 unique proteins
2
1
0
−1
−2
Hepatocyte
Mesenchyme
Immune
Cholangiocyte
Endothelia

Rows in the heatmap correspond to enrichment of transcripts coding for the protein markers included in the Olink panel in respective cell lineages and are scaled (z-score transformation indicating high [red] and low [blue] expression).

[a]At study conception, NASH was the accepted term for this disease. Following a multisociety Delphi consensus initiative, this term has been updated to MASH.[6]

Figure 3. (A)[a] Fazirsiran treatment led to continuous and sustained reductions of proteins in the serum; (B)[b] mapping DEPs to Snucseq data helped to define liver cell origin and contribution of distinct liver cell types to the observed serum DEPs

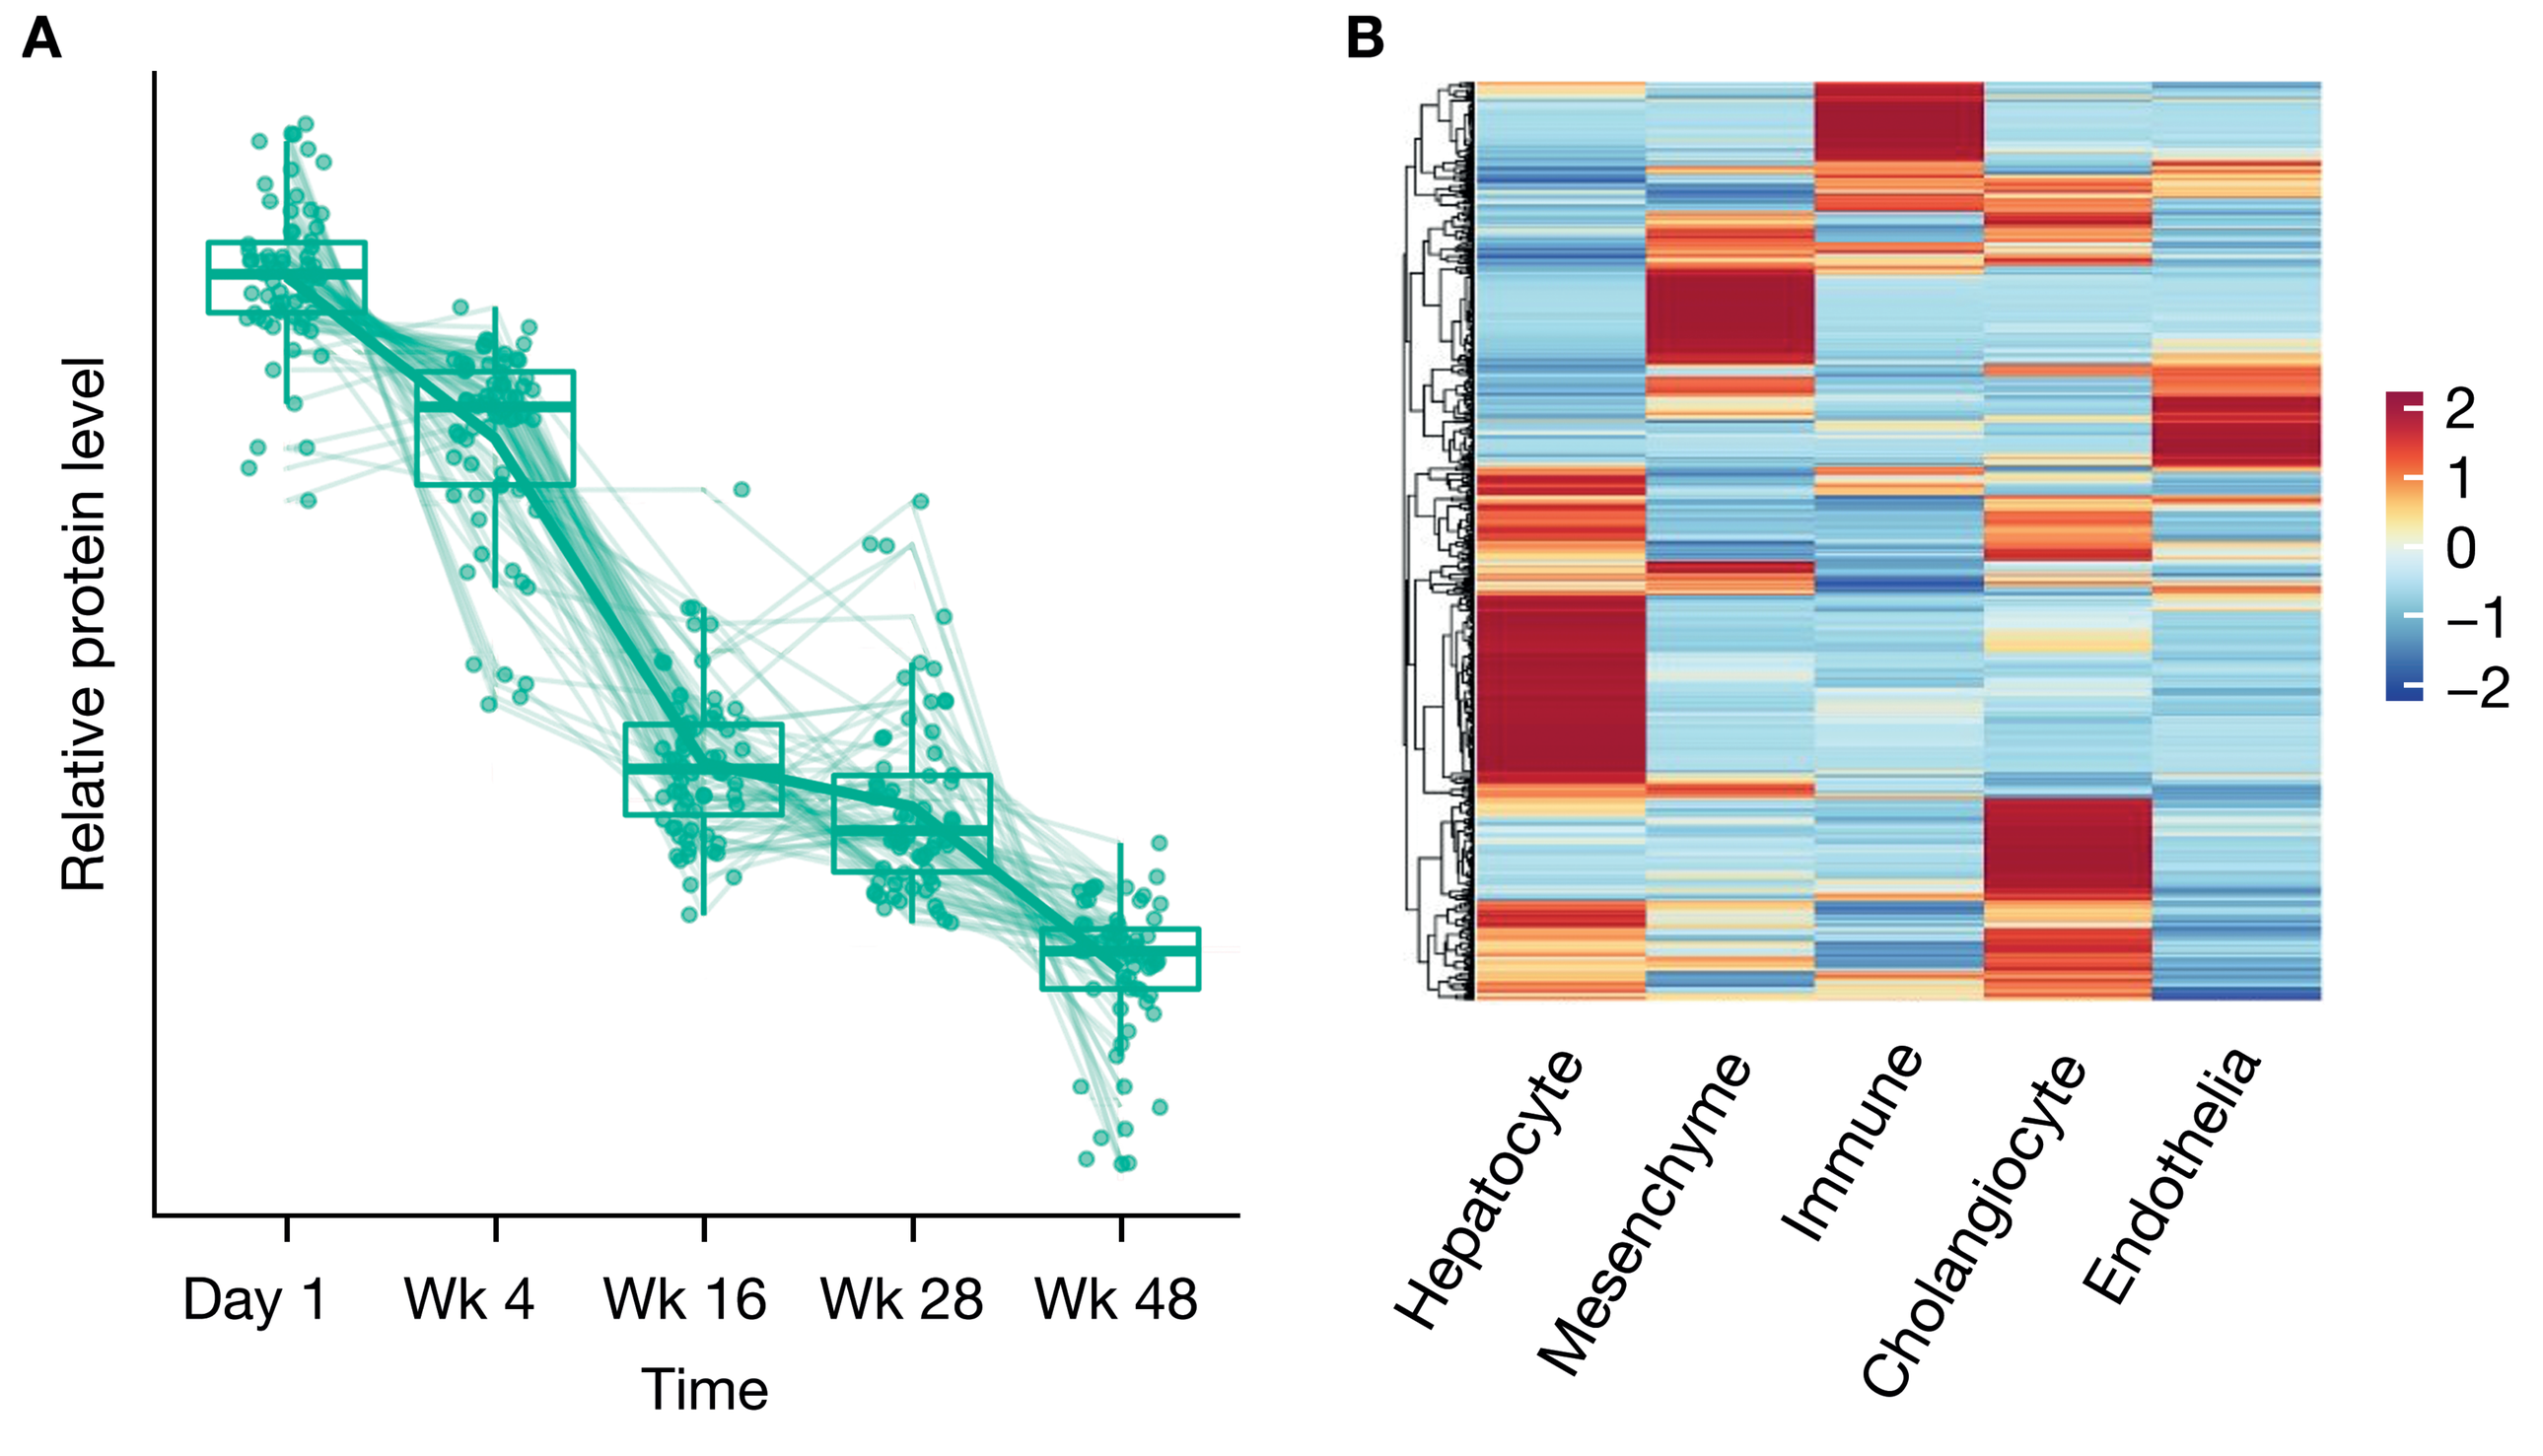
A
B
Relative protein level
Day 1
Wk 4
Wk 16
Wk 28
Wk 48
Time
2
1
0
−1
−2
Hepatocyte
Mesenchyme
Immune
Cholangiocyte
Endothelia

[a]Each circle represents a protein; the box plots shows the median (thick horizontal line), upper and lower interquartile range (thin horizontal lines) and the range (vertical line at each time point) of relative DEP abundance. Baseline samples were collected pre-dose on Day 1.

[b]Rows in the heatmap correspond to enrichment of fazirsiran treatment-responsive proteins in respective cell lineages and are scaled (z-score transformation indicating high [red] and low [blue] expression).

- Ingenuity pathway analysis identified molecular pathways that were improved by fazirsiran treatment (**Figure 5**).
 - Reduced levels of proteins linked to hepatocyte and cell death pathways, including p38 mitogen-activated protein kinase and immunogenic cell death signalling.
 - Decreases in proteins linked to inflammatory response pathways, including Th1/2 activation and cytokine expression; dendritic and natural killer cell crosstalk; macrophages, fibroblasts and endothelial cells; and rheumatoid arthritis-related markers.
 - Reduction in markers associated with liver fibrosis, including moderation of hepatic stellate cell activation, retinoids and wound healing.

Figure 4. Fazirsiran treatment led to significant reductions in circulating protein biomarkers mapped to mesenchymal cells and associated with liver fibrosis and damage

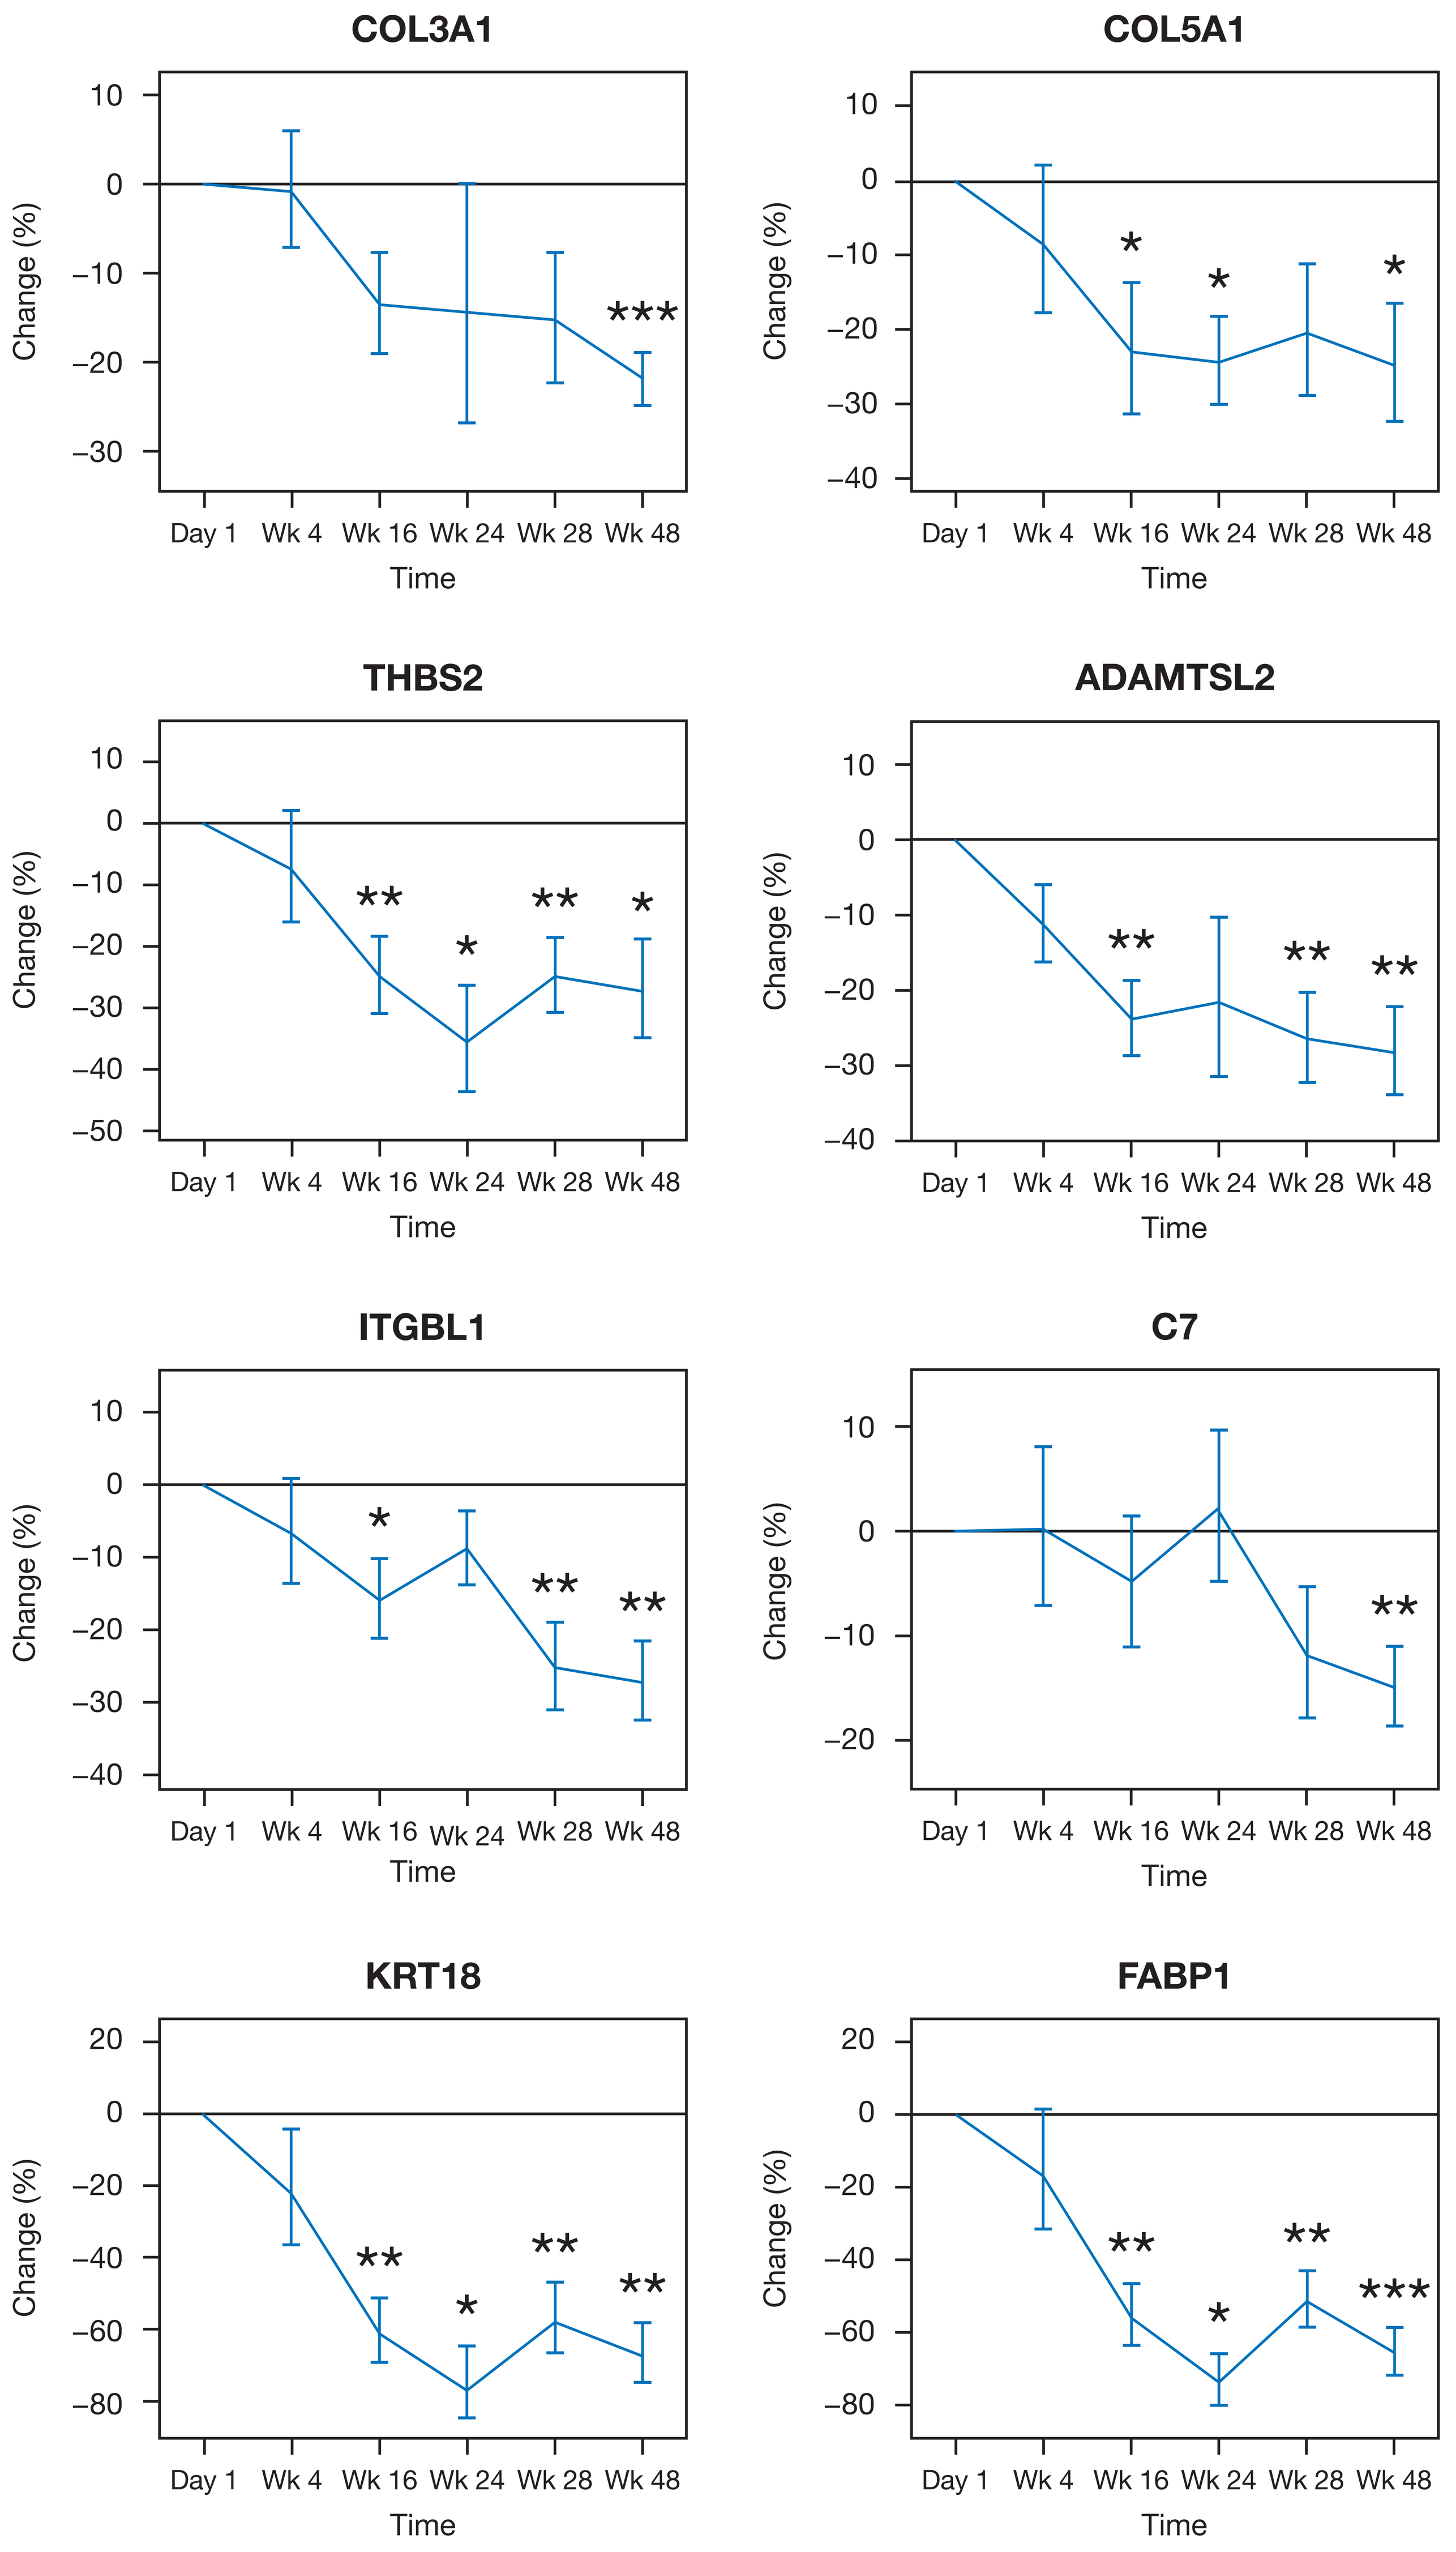
COL3A1
COL5A1
THBS2
ADAMTSL2
ITGBL1
C7
KRT18
FABP1
Change (%)
Day 1
Wk 4
Wk 16
Wk 24
Wk 28
Wk 48
Time

*p < 0.05, **p < 0.01, ***p < 0.001. The mean and standard error are plotted for each time point. Baseline samples were collected pre-dose on Day 1.

Figure 5. Ingenuity pathway analysis showed that fazirsiran-induced proteomic changes from baseline through 48 weeks were linked to the reduced activity of molecular pathways involved in hepatic cell stress and damage, apoptosis, inflammation, extracellular matrix turnover and liver fibrosis

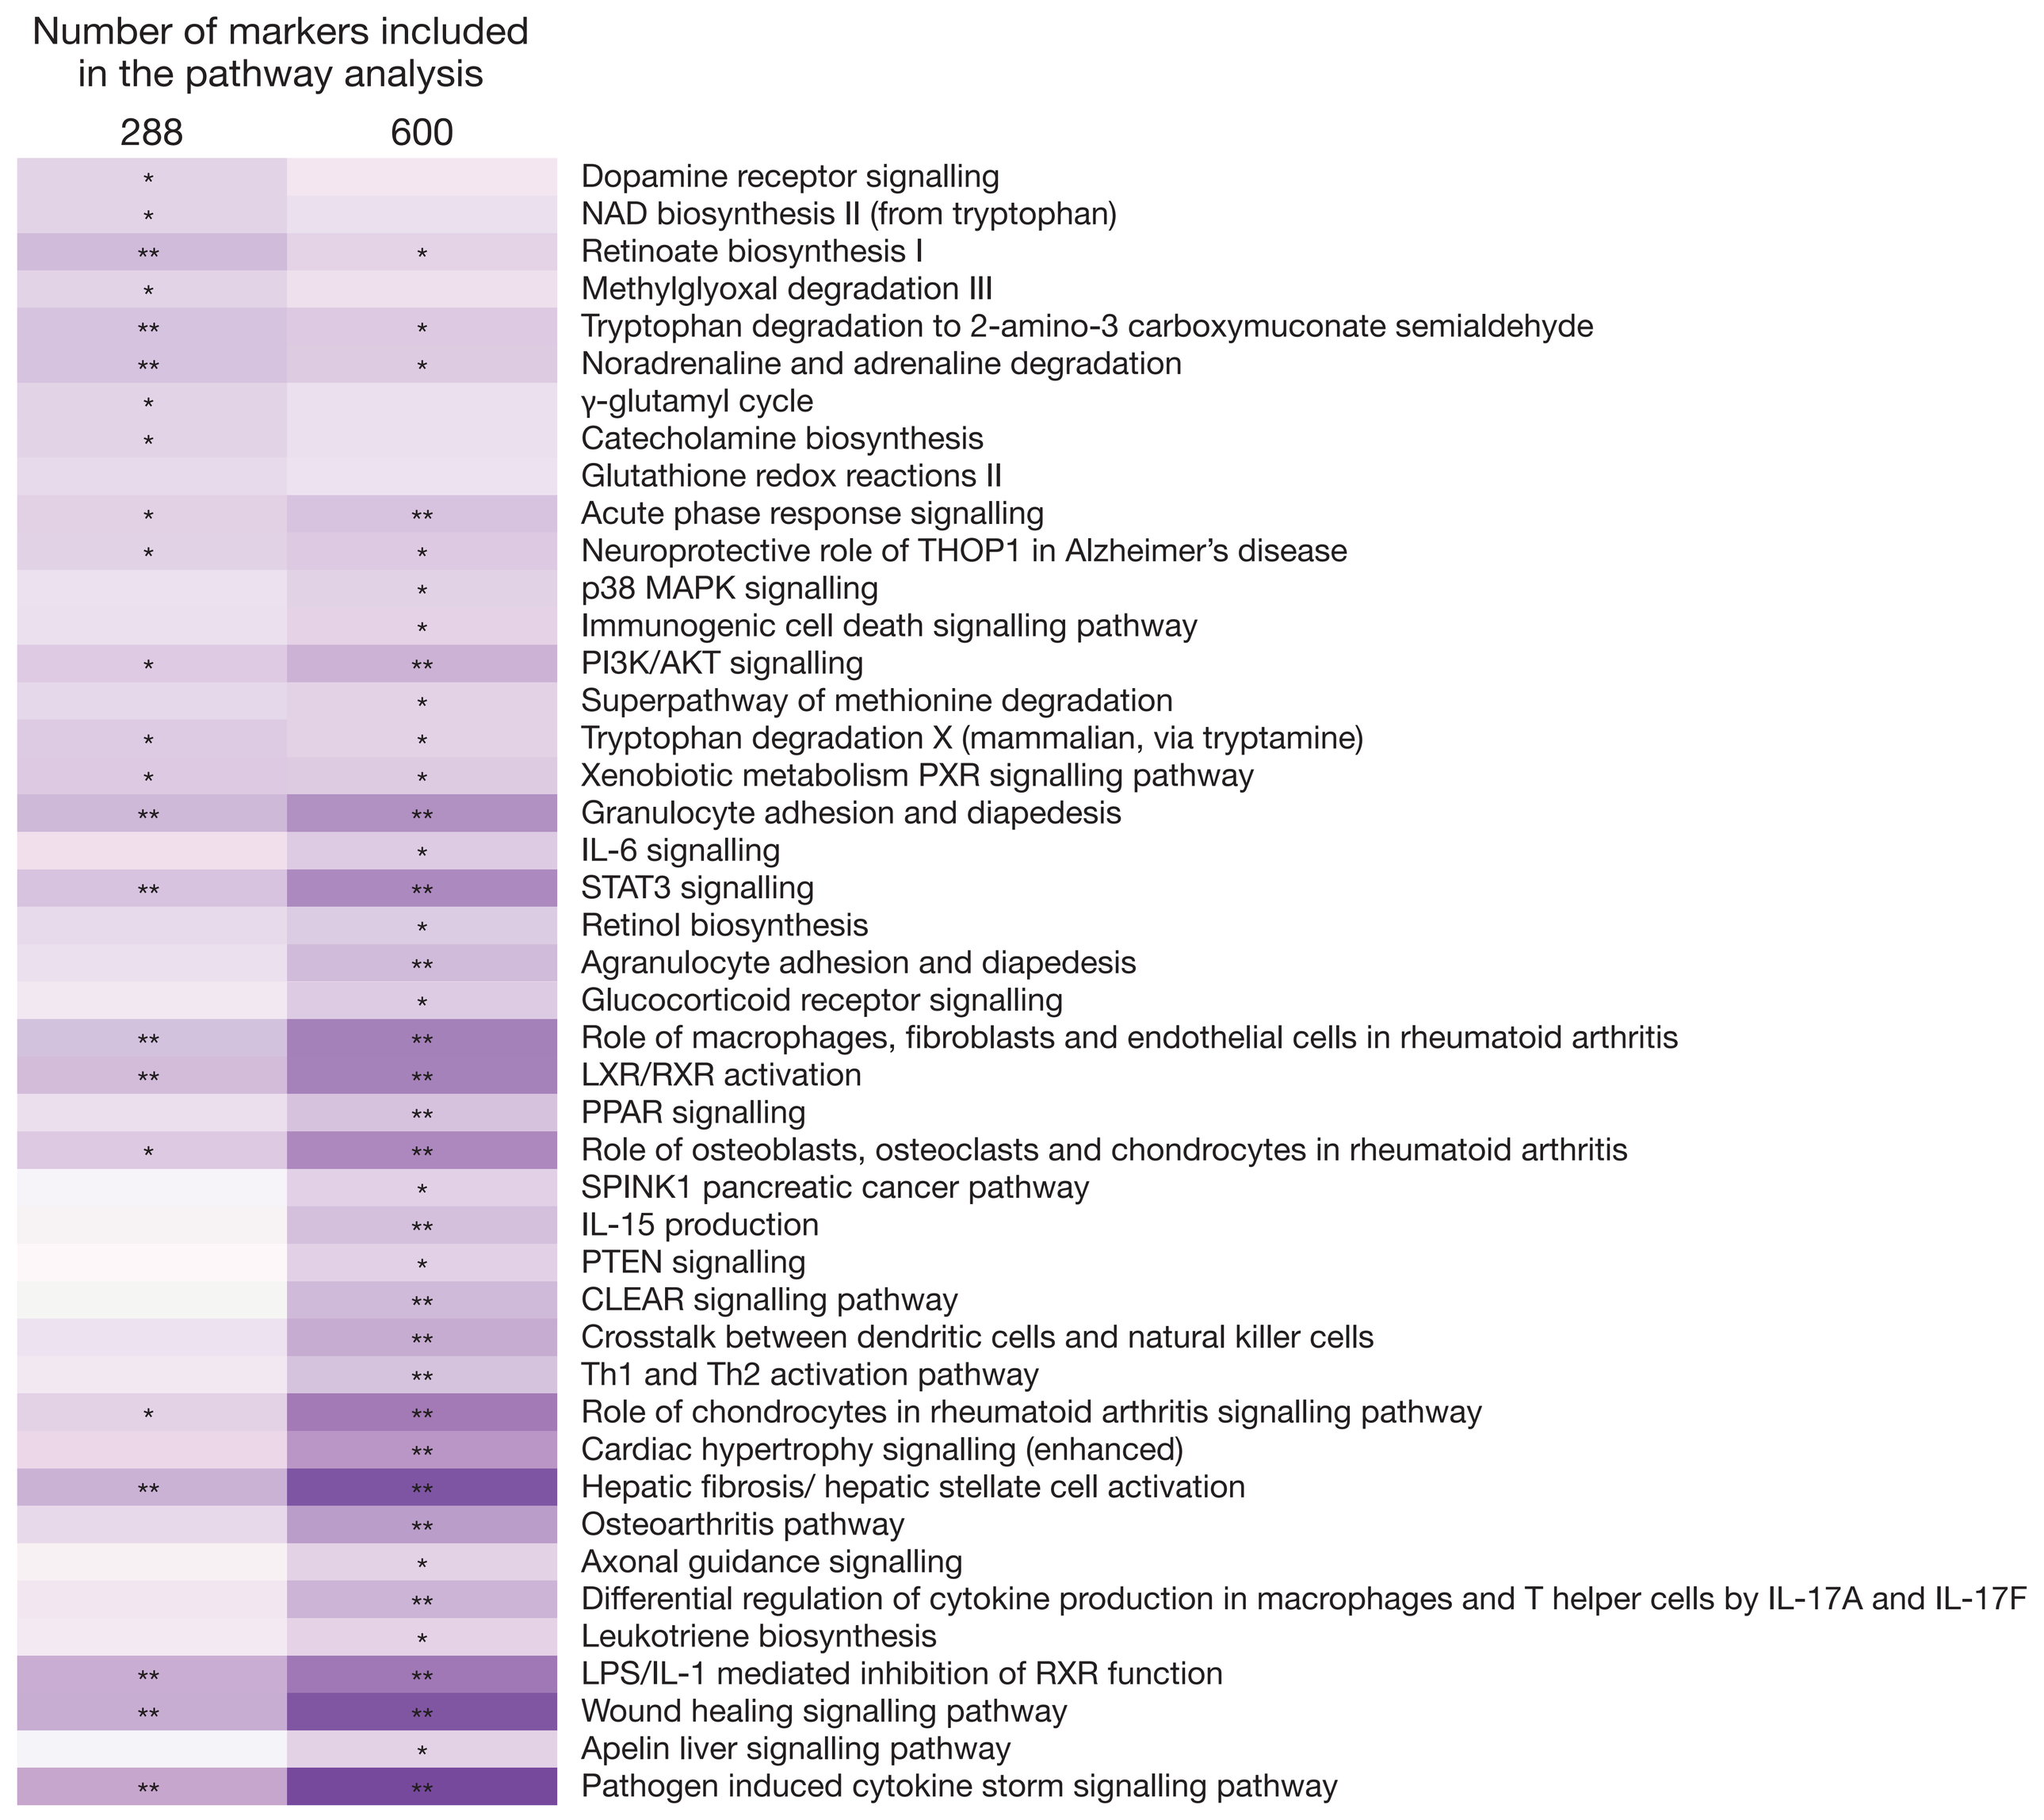
Number of markers included in the pathway analysis
288
600
Dopamine receptor signalling
NAD biosynthesis II (from tryptophan)
Retinoate biosynthesis I
Methylglyoxal degradation III
Tryptophan degradation to 2-amino-3 carboxymuconate semialdehyde
Noradrenaline and adrenaline degradation
γ-glutamyl cycle
Catecholamine biosynthesis
Glutathione redox reactions II
Acute phase response signalling
Neuroprotective role of THOP1 in Alzheimer's disease
p38 MAPK signalling
Immunogenic cell death signalling pathway
PI3K/AKT signalling
Superpathway of methionine degradation
Tryptophan degradation X (mammalian, via tryptamine)
Xenobiotic metabolism PXR signalling pathway
Granulocyte adhesion and diapedesis
IL-6 signalling
STAT3 signalling
Retinol biosynthesis
Agranulocyte adhesion and diapedesis
Glucocorticoid receptor signalling
Role of macrophages, fibroblasts and endothelial cells in rheumatoid arthritis
LXR/RXR activation
PPAR signalling
Role of osteoblasts, osteoclasts and chondrocytes in rheumatoid arthritis
SPINK1 pancreatic cancer pathway
IL-15 production
PTEN signalling
CLEAR signalling pathway
Crosstalk between dendritic cells and natural killer cells
Th1 and Th2 activation pathway
Role of chondrocytes in rheumatoid arthritis signalling pathway
Cardiac hypertrophy signalling (enhanced)
Hepatic fibrosis/ hepatic stellate cell activation
Osteoarthritis pathway
Axonal guidance signalling
Differential regulation of cytokine production in macrophages and T helper cells by IL-17A and IL-17F
Leukotriene biosynthesis
LPS/IL-1 mediated inhibition of RXR function
Wound healing signalling pathway
Apelin liver signalling pathway
Pathogen induced cytokine storm signalling pathway

*p < 0.001, **p < 0.0001.

Conclusions

- This study leveraged a novel proteomic platform for biomarker discovery in patients with AATD-LD, identified candidate biomarkers reflecting disease progression, and demonstrated potential benefit of fazirsiran treatment in reducing cellular stress and damage, apoptosis, inflammation and extracellular matrix turnover/fibrosis.
- Collectively, these findings provide molecular evidence to support potential clinical benefit of fazirsiran in patients with AATD-LD.
- Biomarkers identified in these analyses may have clinical utility but require validation in larger studies.
- The study was limited by:
 - the small sample size (n = 16)
 - the lack of comparator data from healthy patients or patients who received placebo; future research should generate additional Olink proteomic data from these comparator populations.
- Quantitative assays, such as enzyme-linked immunosorbent assays, would prove beneficial in validating findings from this study and bridging the gap to clinical practice.

Abbreviations

ADAMTSL2, disintegrin and metalloproteinase domain with thrombospondin motif-like protein 2; C7, complement component 7; CLEAR, coordinate lysosomal expression and regulation; COL3A1, collagen type III alpha 1 chain; COL5A1, collagen type V alpha 1 chain; DEP, differentially expressed protein; FABP1, fatty acid binding protein 1; FDR, false discovery rate; IL-6, interleukin 6; IL-15, interleukin 15; IL-17A, interleukin 17A; IL-17F, interleukin 17F; ITGBL1, integrin subunit beta like 1; KRT18, type I intermediate filament chain keratin 18; LPS/IL-1, lipopolysaccharide/interleukin 1; LXR/RXR, liver X receptor/retinoid X receptor; MAPK, mitogen-activated protein kinase; MASH, metabolic dysfunction-associated steatohepatitis; NAD, nicotinamide adenine dinucleotide; NASH, non-alcoholic steatohepatitis; PI3K/AKT, phosphoinositide-3-kinase/protein kinase B; PPAR, peroxisome proliferator-activated receptor; PTEN, phosphatase and tensin homolog; PXR, pregnane X receptor; Snucseq, single cell nucleus RNA-sequencing; SPINK1, serine protease inhibitor kazal type 1; STAT3, signal transducer and activator of transcription 3; Th1, type 1 T helper; Th2, type 2 T helper; THBS2, thrombospondin 2; THOP1, thimet oligopeptidase; UMAP, uniform manifold approximation and projection for dimension reduction; Wk, week.

Acknowledgements

We wish to thank all the patients who participated in the studies. Medical writing support was provided by Rebecca Tooze, PhD, of Oxford PharmaGenesis, Oxford, UK and funded by Takeda Development Center Americas, Inc.

Funding

This study was funded by Takeda Development Center Americas, Inc.

References

References can be found in the supplementary material accessed via the QR code.

Disclosures

J-CC, **RH**, **FH**, **JC**, **MDP**, **NKD** and **PT** are employees and stockholders of Takeda Development Center Americas, Inc. **TS** is an employee of Arrowhead Pharmaceuticals. **PS** has received grant support and lecture fees from CSL Behring; grant support from Arrowhead Pharmaceuticals, Dicerna Pharmaceuticals and Vertex Pharmaceuticals; advisory board/consulting fees from BioMarin Pharmaceutical, BridgeBio, GlaxoSmithKline, Intellia Pharmaceuticals and Takeda Pharmaceuticals.

Presented at the European Association for the Study of the Liver (EASL) International Liver Congress 2024, 5–8 June 2024, Milan, Italy